Abstract #2812

Margetuximab plus Pembrolizumab for Treatment of Patients with HER2-Positive Gastroesophageal Adenocarcinoma (GEA) Post-Trastuzumab: Survival Analysis

D.V.T. Catenacci[1], H. Park[2], H.E. Uronis[3], Y.-K. Kang[4], M. Ng[5], P.C. Enzinger[6], K.-W. Lee[7], K.H. Lim[2], P.J. Gold[8], J. Lacy[9], S.H. Park[10], K. Huber[11], A. Wynter-Horton[11], J. Nordstrom[11], Y. Yang[11], P. Moore[11], D. Li[11], T. Wu[11], J. Wigginton[11], J. Baughman[11], M. Rosales[11], J. Davidson-Moncada[11], Y. J. Bang[12]

[1]The University of Chicago Medical Center, Chicago, IL, USA; [2]Washington University, St. Louis, MO, USA; [3]Duke University Medical Center, Durham, NC, USA; [4]Asan Medical Center, University of Ulsan College of Medicine, Seoul, KR; [5]National Cancer Centre, Singapore; [6]Dana-Farber Cancer Institute, Boston, MA, USA; [7]Seoul National University Bundang Hospital, Seoul, KR; [8]Swedish Cancer Institute, Seattle, WA, USA; [9]Yale Cancer Center, New Haven, CT, USA; [10]Samsung Medical Center, Seoul, KR; [11]MacroGenics, Inc., Rockville, MD, USA; [12]Seoul National University Hospital, Seoul, KR

NCT02689284

Background

- Trastuzumab + chemotherapy is standard treatment in 1st line advanced HER2+ gastroesophageal adenocarcinoma (GEA); however, patients tend to progress in 6–8 months
- Consistent with previous literature, we have noted ~40% of patients have a loss of HER2 expression post trastuzumab (abstract #2794), likely underlying the lack of efficacy anti-HER2 agents in prior 2nd line studies
- Margetuximab is an investigational next generation anti-HER2 monoclonal antibody with an engineered Fc domain that confers enhanced Fc-dependent antitumor activities across all FcγRIIIA (CD16A) genotypes
- Margetuximab has demonstrated single agent antitumor activity in patients with HER2+ GEA in a Phase 1 study
- Current standard of care in 2nd line GEA, ramucirumab + taxane, has median progression-free survival (mPFS) of 4.4 months and median overall survival (mOS) of 9.6 months
- Chemotherapy-backbone HER2-targeted agents tested in 2nd line HER2+ GEA TyTAN and GATSBY studies showed mPFS and mOS ranged 2.4–5.4 months, and 7.1–11.2 months, respectively
- Pembrolizumab, in 2nd line KN061 study, showed mPFS and mOS of 1.5 months and 9.1 months in GEA patients with PD-L1 CPS >1, and both were lower for PD-L1 all comers
- Herein we present survival analysis of an ongoing study in patients receiving margetuximab plus pembrolizumab at the recommended Phase 2 dose (RP2D), 15 mg/kg of margetuximab and 200 mg of pembrolizumab q3wk, a chemotherapy-free treatment, in HER2+ GEA patients post trastuzumab

Methods

- HER2-positive (archival IHC3+, or ICH2+/FISH positive), PD-L1 unselected GEA patients (data cut 10 July 2019)
- Median PFS and OS analysis performed on patients post progression on trastuzumab-based therapy
 - 92 patients treated at RP2D of 15 mg/kg margetuximab + 200 mg pembrolizumab included in analysis (data cut 10 July 2019)
 - Biomarker subgroups including archival PD-L1 and HER2 IHC, ERRB2 ctDNA (prior to 2nd line therapy), and tumor site (GC vs GEJ)

Margetuximab: Fc-engineered to Activate Immune Responses

Trastuzumab

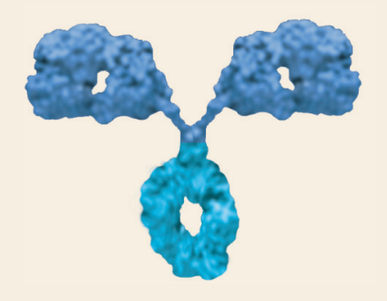

Fab:
- Binds HER2 with high specificity
- Disrupts signaling that drives cell proliferation and survival

Fc:
- Wild-type immunoglobulin G1 (IgG1) immune effector domains
- Binds and activates immune cells

Margetuximab[1,2]

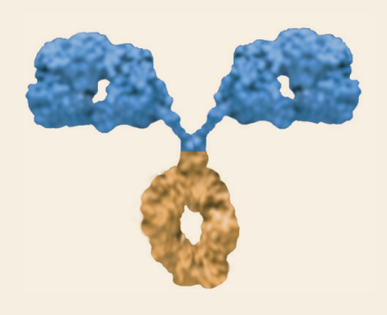

Fab:
- Same specificity and affinity
- Similarly disrupts signaling

Fc engineering:
- ↑ Affinity for activating FcγRIIIA (CD16A)
- ↓ Affinity for inhibitory FcγRIIB (CD32B)

Margetuximab Binding to FcγR Variants

Receptor Type	Receptor	Allelic Variant	Relative Fc Binding	Affinity Fold-Change
Activating	CD16A	158F	Lower	6.6 x ↑
		158V	Higher	4.7 x ↑
	CD32A	131R	Lower	6.1 x ↓
		131H	Higher	↔
Inhibitory	CD32B	232I/T	Equivalent	8.4 x ↓

[1]Nordstrom JL, et al. *Breast Cancer Res.* 2011;13(6):R123. [2]Stavenhagen JB, et al. *Cancer Res.* 2007;67(18):8882-8890.

Margetuximab Enhances Innate Immunity In Vitro

Greater relative cytotoxicity of margetuximab with NK cells from CD16A-158F allele carriers

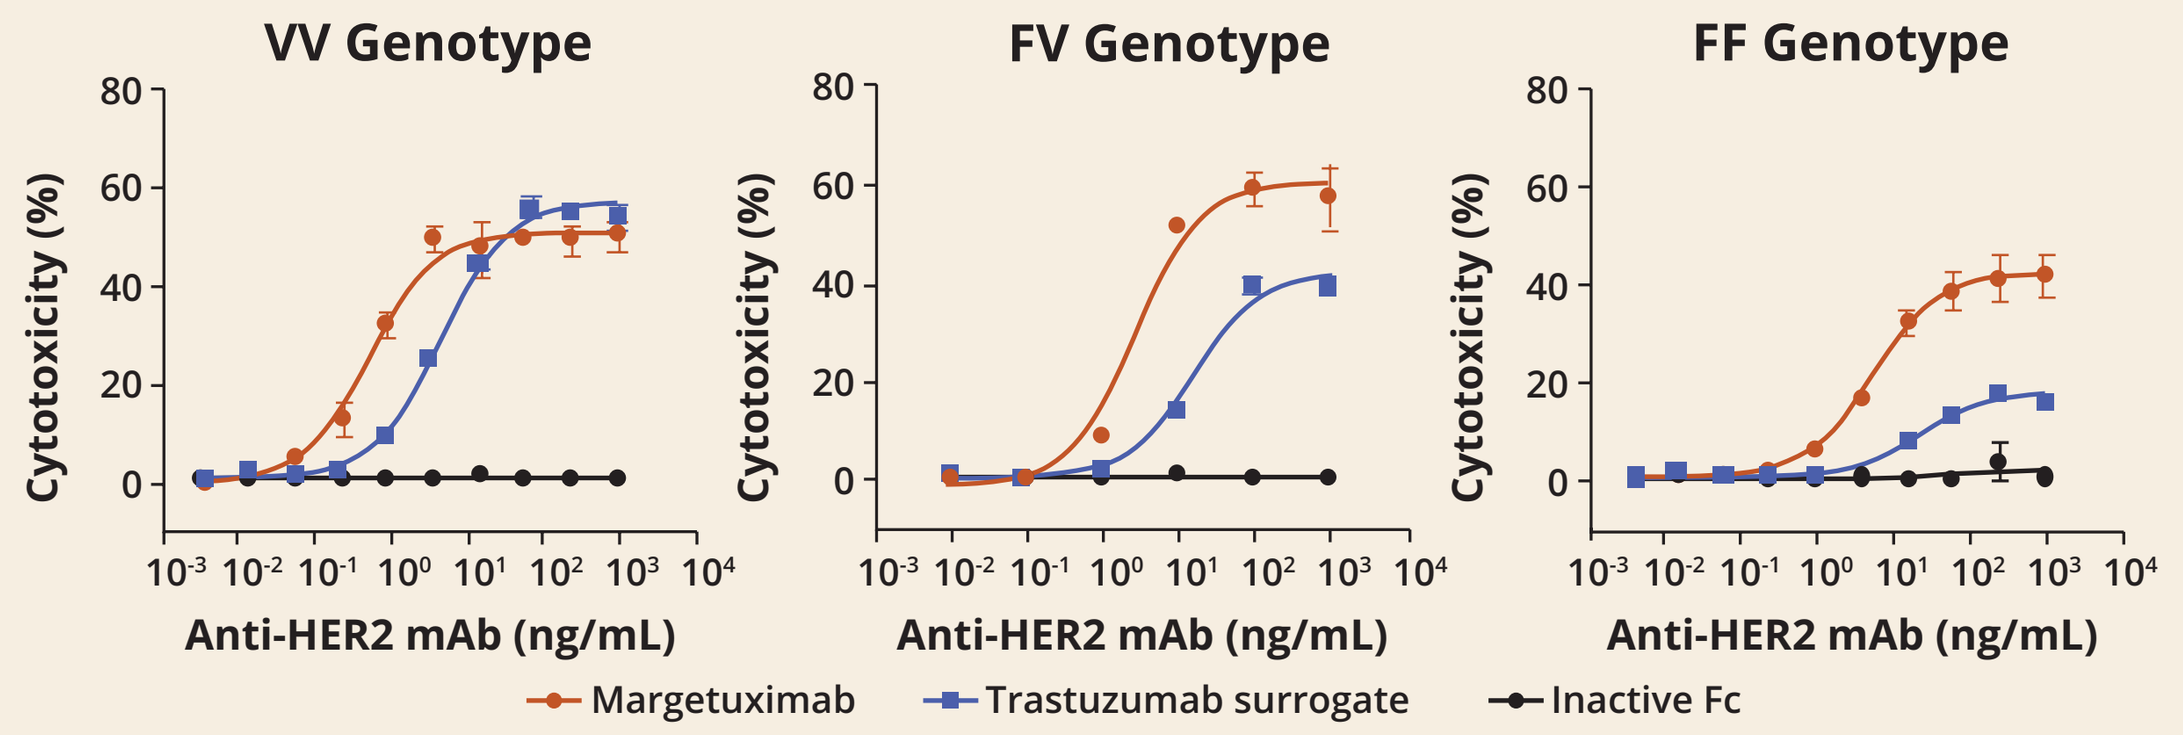

VV Genotype
FV Genotype
FF Genotype
Cytotoxicity (%)
Anti-HER2 mAb (ng/mL)
Margetuximab
Trastuzumab surrogate
Inactive Fc

Preclinical Assay of Antibody-Dependent Cellular Cytotoxicity (ADCC)[1]

- **Effector Cells:** Human NK cells from donors with CD16A genotypes 158VV, 158FV, and 158FF
- **Target Cells:** JIMT-1 HER2+ breast cancer cell line resistant to trastuzumab antiproliferative activity
- **Cellular Assay:** 3:1 Effector:Target ratio; 24-hour incubation time; endpoint: % lactate dehydrogenase release

[1]Nordstrom JL, et al. Breast Cancer Res. 2011;13(6):R123. mAb: monoclonal antibody; NK: natural killer.

Anti-PD-1 Enhances Margetuximab-mediated NK Cell Cytolytic Potential In Vitro

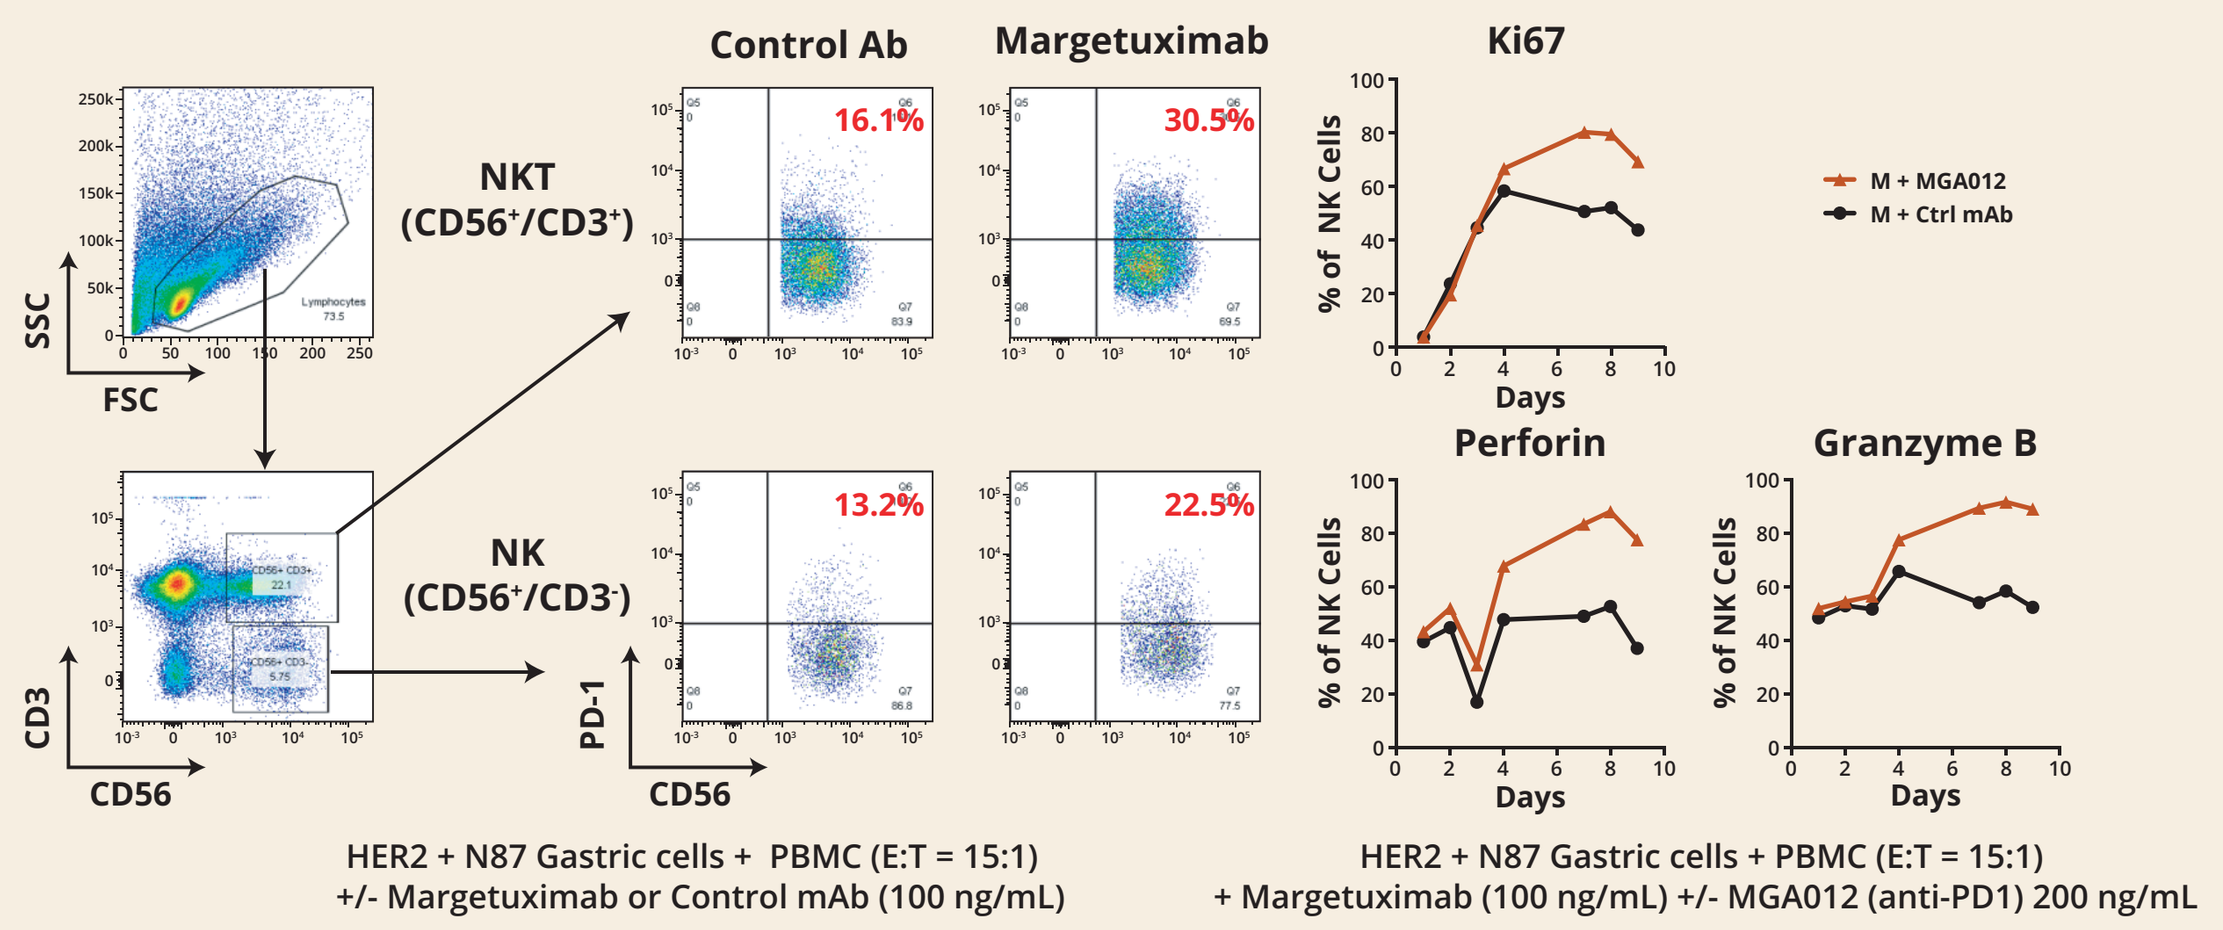

Margetuximab Induces PD-1 Expression on NK and NKT Cells
Anti-PD-1 Enhances Margetuximab-mediated NK Cell Proliferation and Expression of Granzyme B/Perforin
Control Ab
Margetuximab
Ki67
NKT (CD56+/CD3+)
NK (CD56+/CD3-)
16.1%
30.5%
13.2%
22.5%
SSC
FSC
CD3
CD56
PD-1
% of NK Cells
Days
Perforin
Granzyme B
M + MGA012
M + Ctrl mAb
HER2 + N87 Gastric cells + PBMC (E:T = 15:1) +/- Margetuximab or Control mAb (100 ng/mL)
HER2 + N87 Gastric cells + PBMC (E:T = 15:1) + Margetuximab (100 ng/mL) +/- MGA012 (anti-PD1) 200 ng/mL

Proposed Margetuximab and Pembrolizumab Synergistic Mechanisms of Action

Margetuximab engages the innate immune system and activates the adaptive immune system supporting combination with checkpoint inhibitors

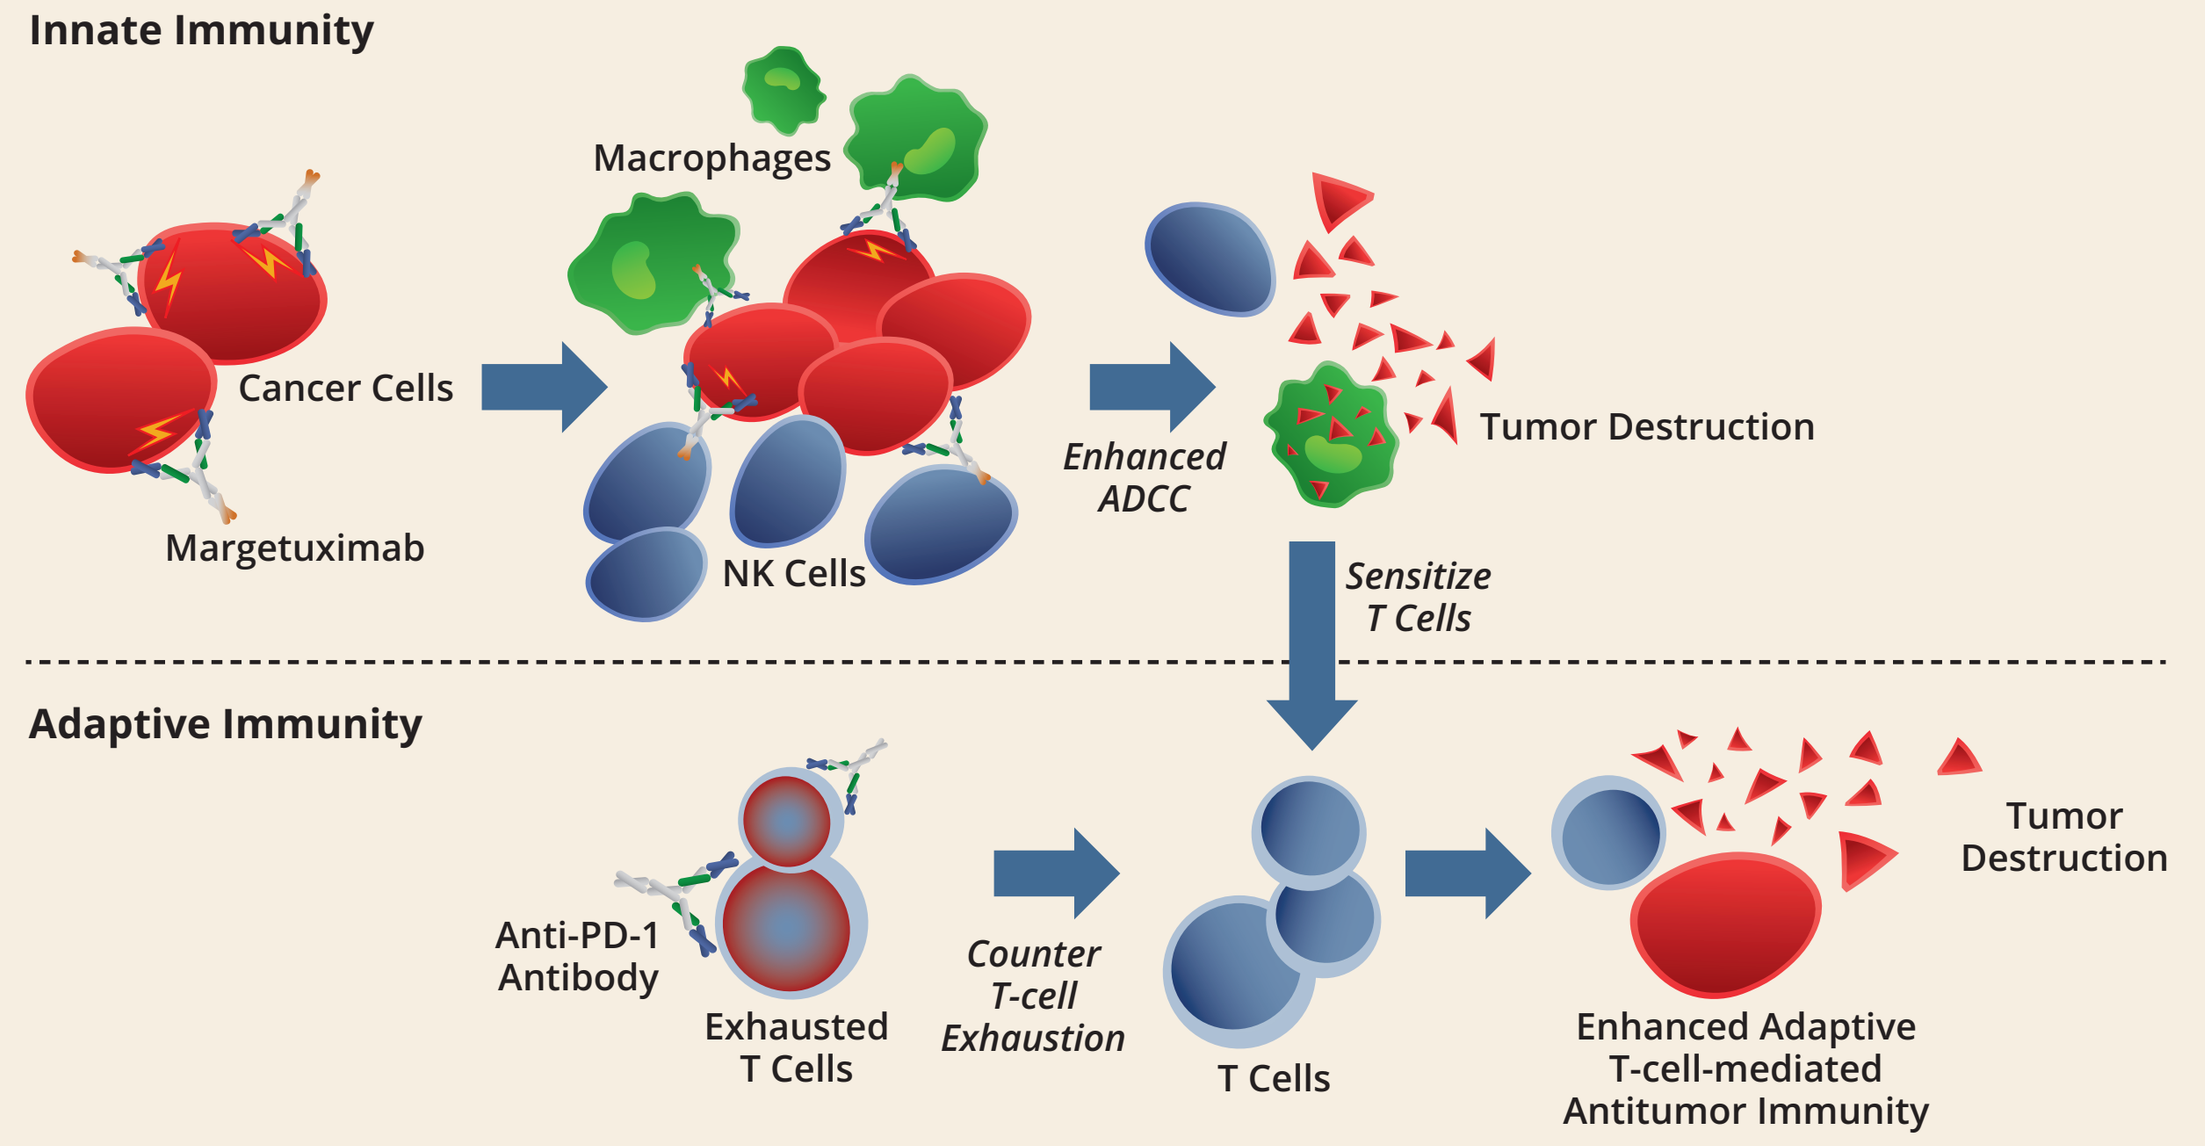

Innate Immunity
Macrophages
Cancer Cells
Margetuximab
NK Cells
Enhanced ADCC
Tumor Destruction
Sensitize T Cells
Adaptive Immunity
Anti-PD-1 Antibody
Exhausted T Cells
Counter T-cell Exhaustion
T Cells
Tumor Destruction
Enhanced Adaptive T-cell-mediated Antitumor Immunity

Study Design

Fully Enrolled Phase 2 Study in Advanced HER2+ Gastric Carcinoma

Dose Escalation	Dose Expansion #1	Dose Expansion #2
(n=3–6 per margetuximab dose)	(margetuximab 15 mg/kg q3W + pembrolizumab 200 mg q3W)	(margetuximab 15 mg/kg q3W + pembrolizumab 200 mg q3W)
Margetuximab 10 mg/kg (n=3), 15 mg/kg (n=6) q3w + pembrolizumab 200 mg q3w	Gastric and Gastroesophageal (n=60)	Gastric (HER2 3+) (n=25)

Patient Characteristics

Characteristic		All Patients (n=92)
Age	Mean ± SD	60.2 ± 12.83
	Median (Range)	61.0 (19, 85)
Gender [n (%)]	Male	75 (81.5)
	Female	17 (18.5)
Race [n (%)]	Asian	51 (55.4)
	White	34 (37.0)
	Other	4 (4.3)
	Black or African American	3 (3.3)
ECOG Status [n (%)]	0	33 (35.9)
	1	59 (64.1)
Diagnosis [n (%)]	Gastric Cancer	61 (66.3)
	GEJ Cancer	31 (33.7)
Microsatellite Stable [n (%)]		84 (91.3)

Results

Progression-free Survival in Overall Population

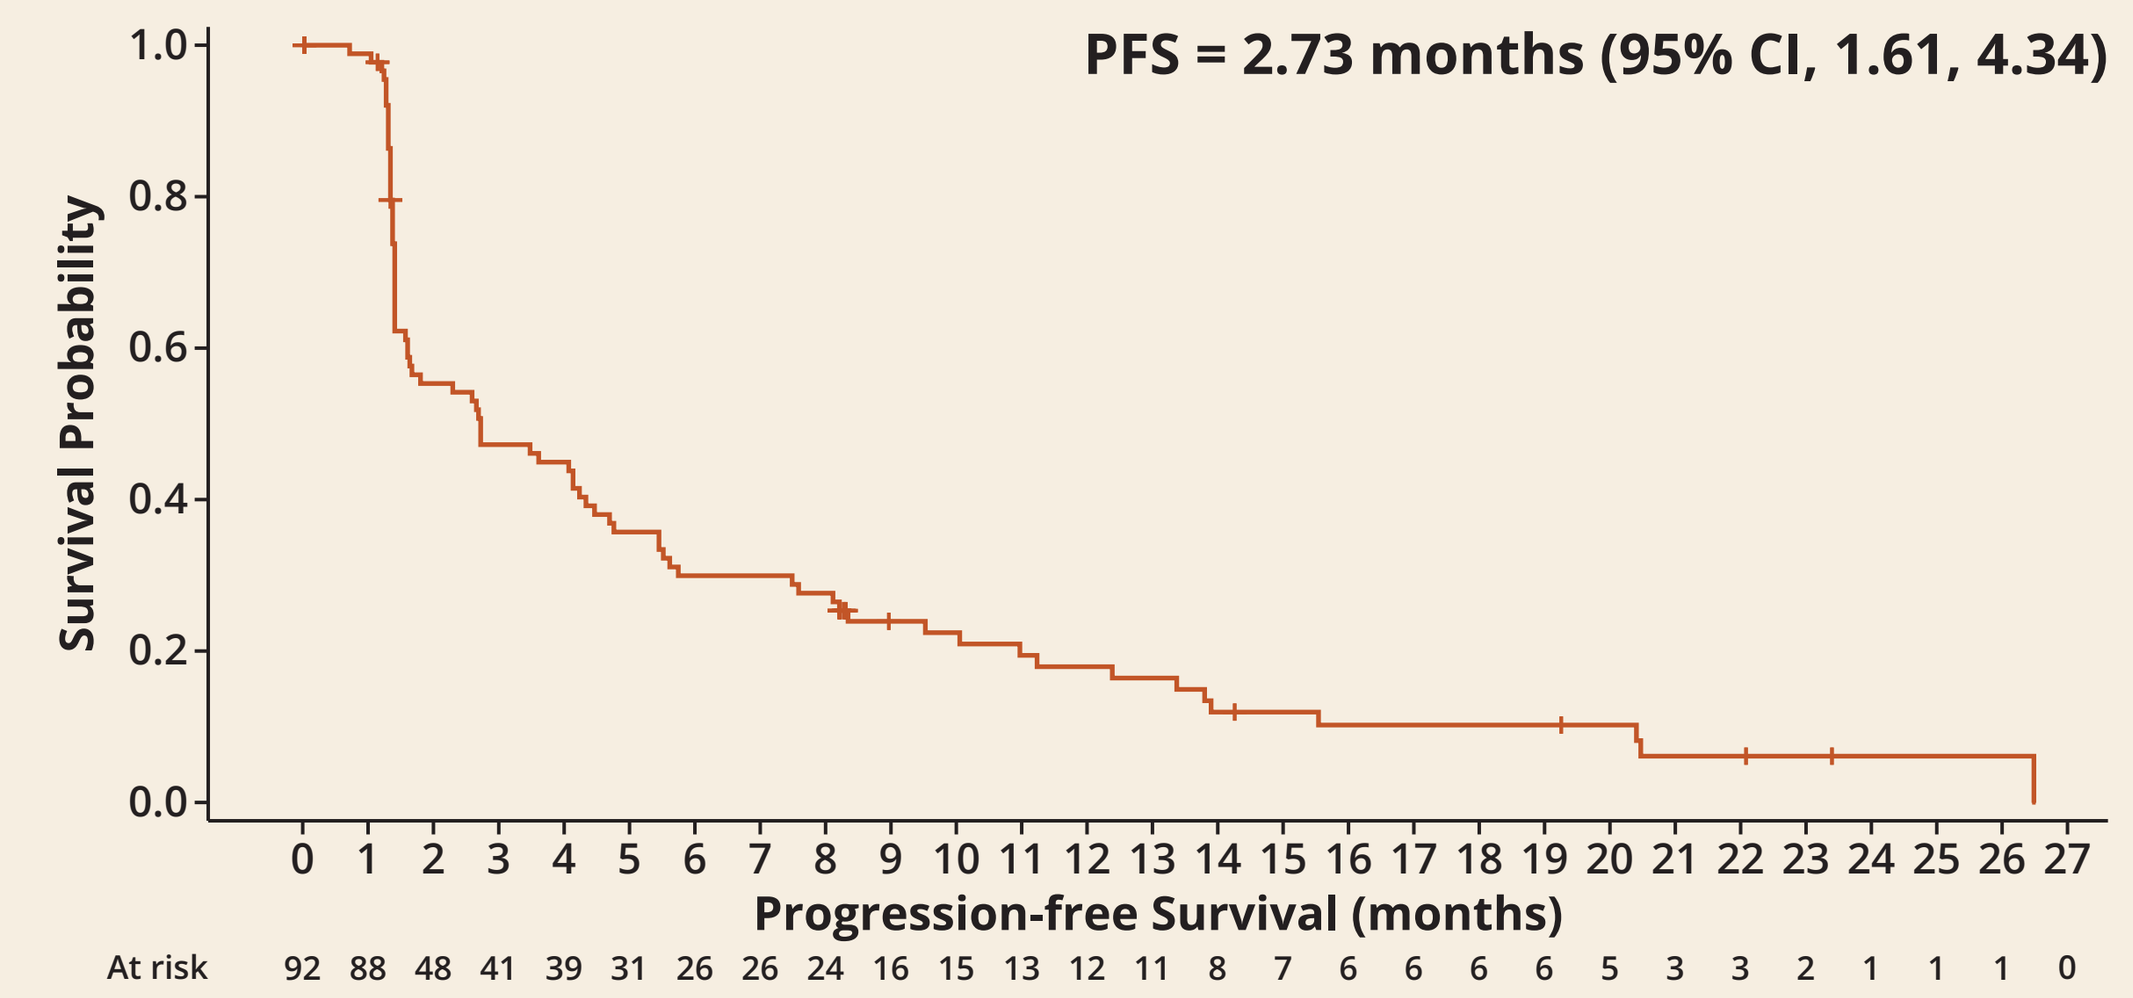

PFS = 2.73 months (95% CI, 1.61, 4.34)
Survival Probability
Progression-free Survival (months)
At risk 92 88 48 41 39 31 26 26 24 16 15 13 12 11 8 7 6 6 6 6 5 3 3 2 1 1 1 0

Survival in Overall Population

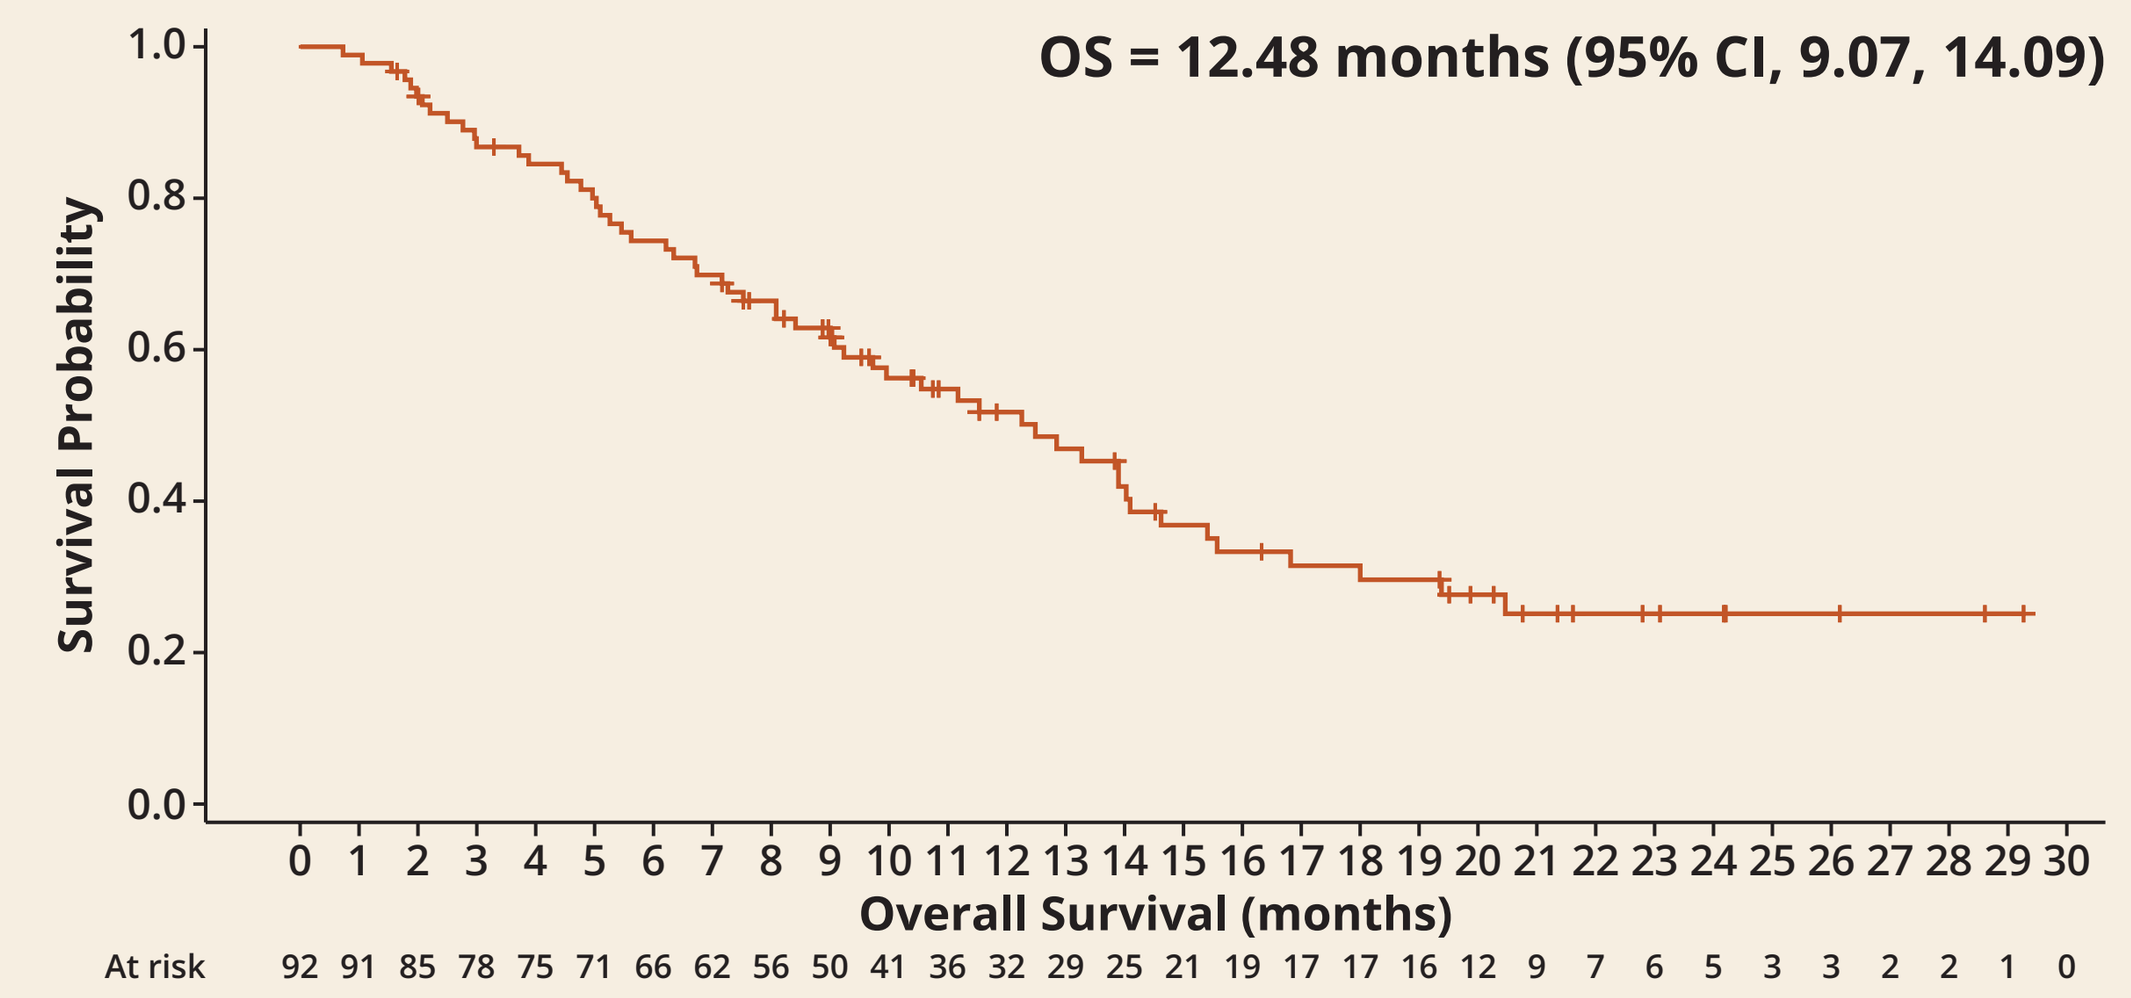

OS = 12.48 months (95% CI, 9.07, 14.09)
Survival Probability
Overall Survival (months)
At risk 92 91 85 78 75 71 66 62 56 50 41 36 32 29 25 21 19 17 17 16 12 9 7 6 5 3 3 2 2 1 0

Progression-Free Survival by Tumor Site

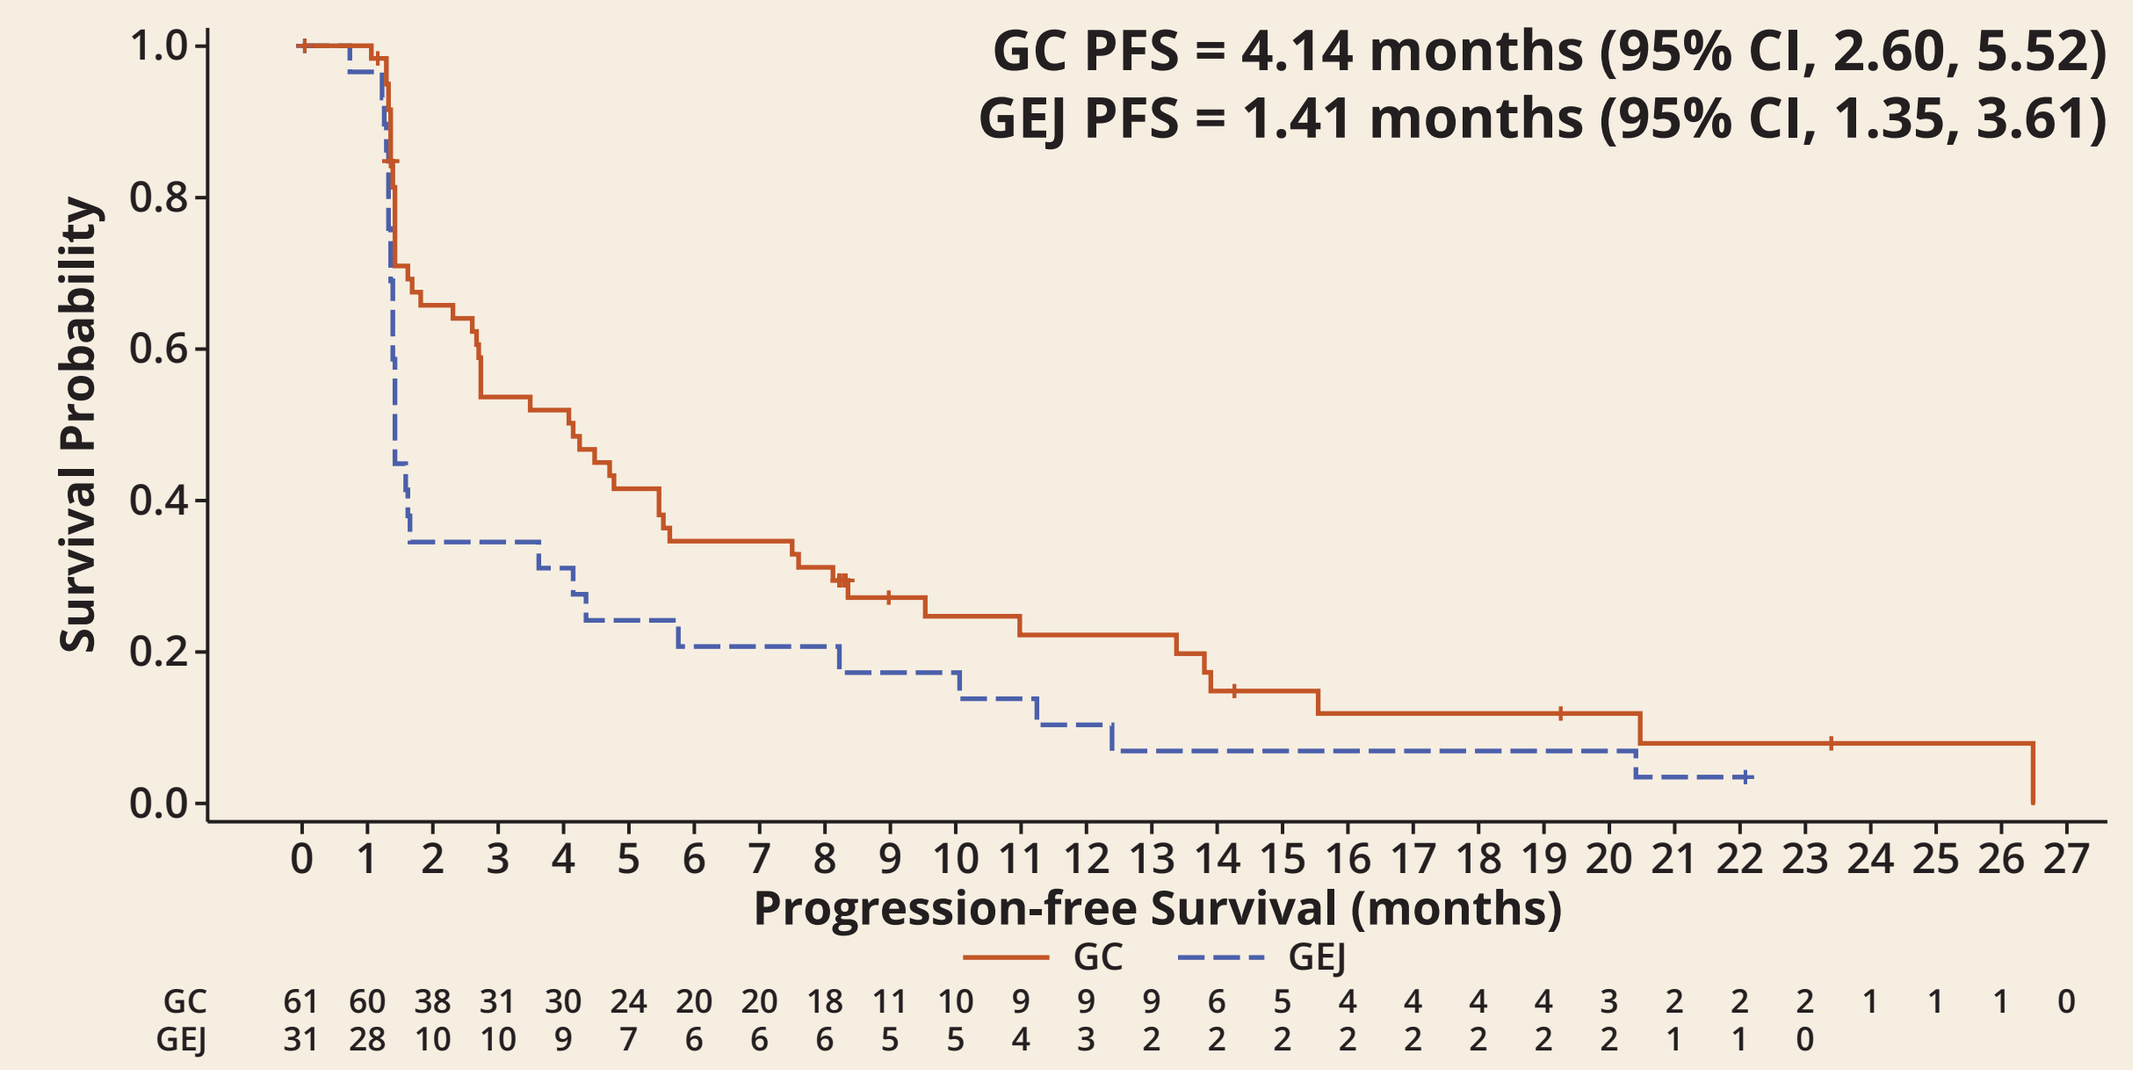

GC PFS = 4.14 months (95% CI, 2.60, 5.52)
GEJ PFS = 1.41 months (95% CI, 1.35, 3.61)
Survival Probability
Progression-free Survival (months)
GC
GEJ
GC 61 60 38 31 30 24 20 20 18 11 10 9 9 9 6 5 4 4 4 4 3 2 2 2 1 1 1 0
GEJ 31 28 10 10 9 7 6 6 6 5 5 4 3 2 2 2 2 2 2 2 2 1 1 0

Overall Survival by Tumor Site

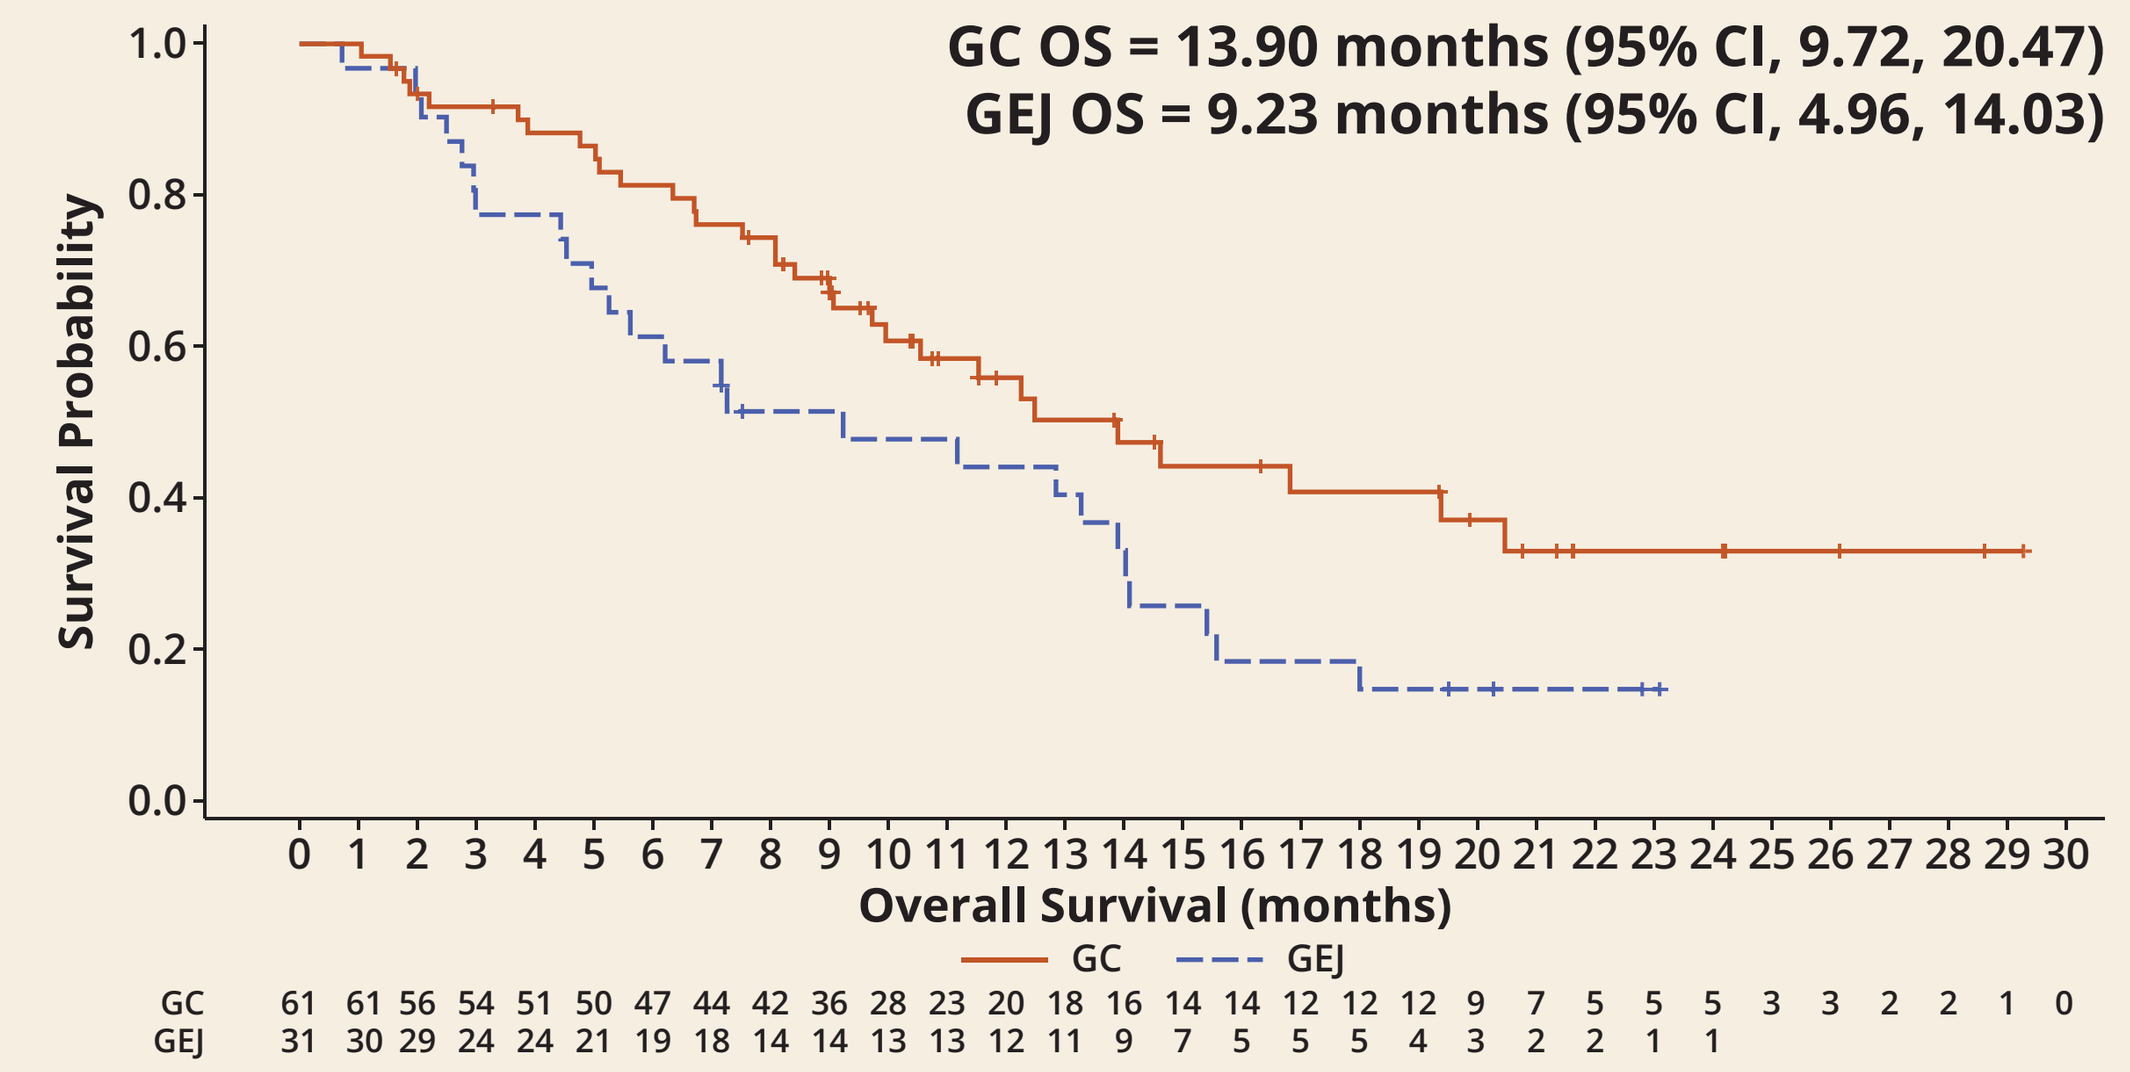

GC OS = 13.90 months (95% CI, 9.72, 20.47)
GEJ OS = 9.23 months (95% CI, 4.96, 14.03)
Survival Probability
Overall Survival (months)
GC
GEJ
GC 61 61 56 54 51 50 47 44 42 36 28 23 20 18 16 14 14 12 12 12 9 7 5 5 5 3 3 2 2 1 0
GEJ 31 30 29 24 24 21 19 18 14 14 13 13 12 11 9 7 5 5 5 4 3 2 2 1 1

Enhancement of PFS in HER2 (IHC3+) and PD-L1+ Population

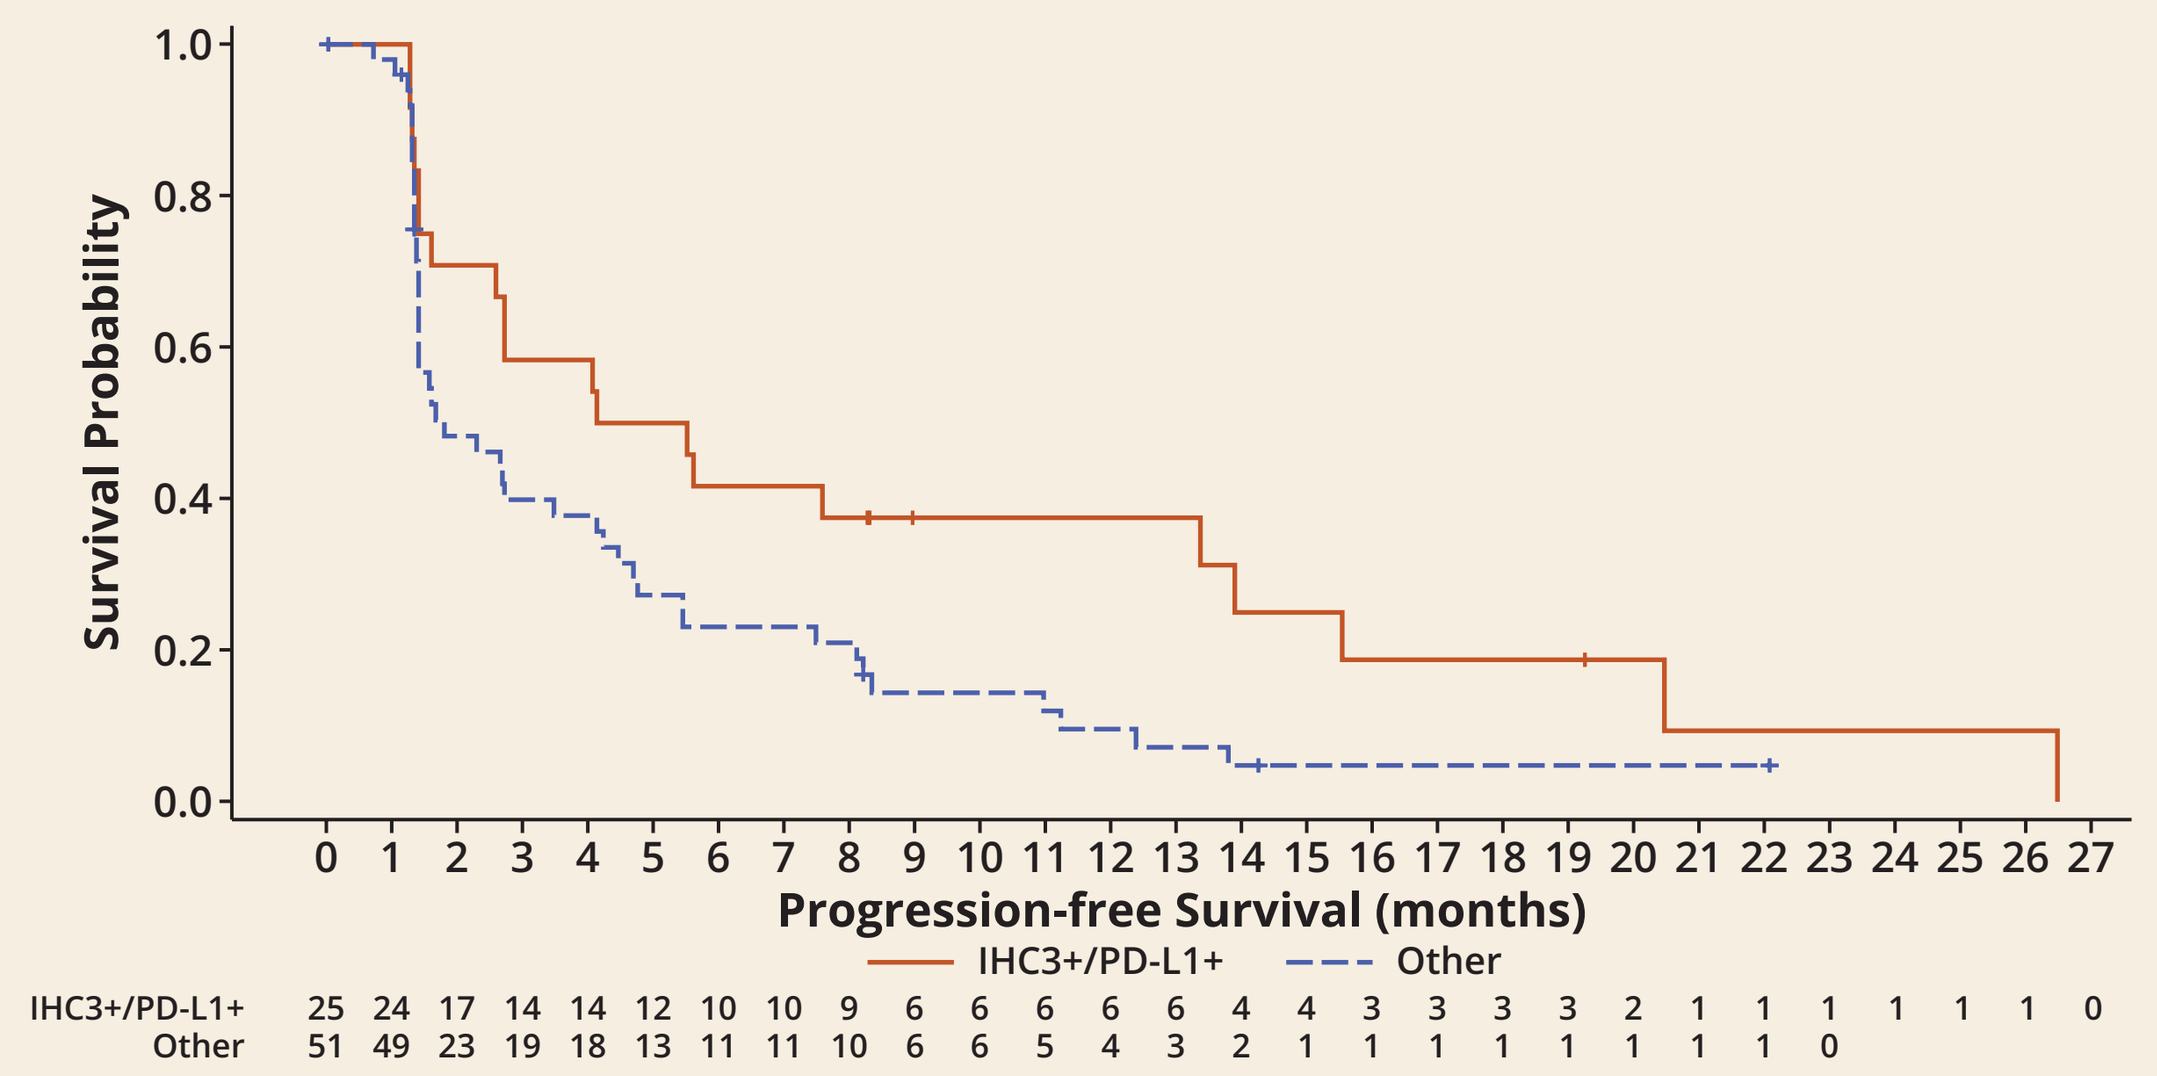

Survival Probability
Progression-free Survival (months)
IHC3+/PD-L1+
Other
IHC3+/PD-L1+ 25 24 17 14 14 12 10 10 9 6 6 6 6 6 4 4 3 3 3 3 2 1 1 1 1 1 1 0
Other 51 49 23 19 18 13 11 11 10 6 6 5 4 3 2 1 1 1 1 1 1 1 1 0

Enhancement of OS in HER2 (IHC3+) and PD-L1+ Population

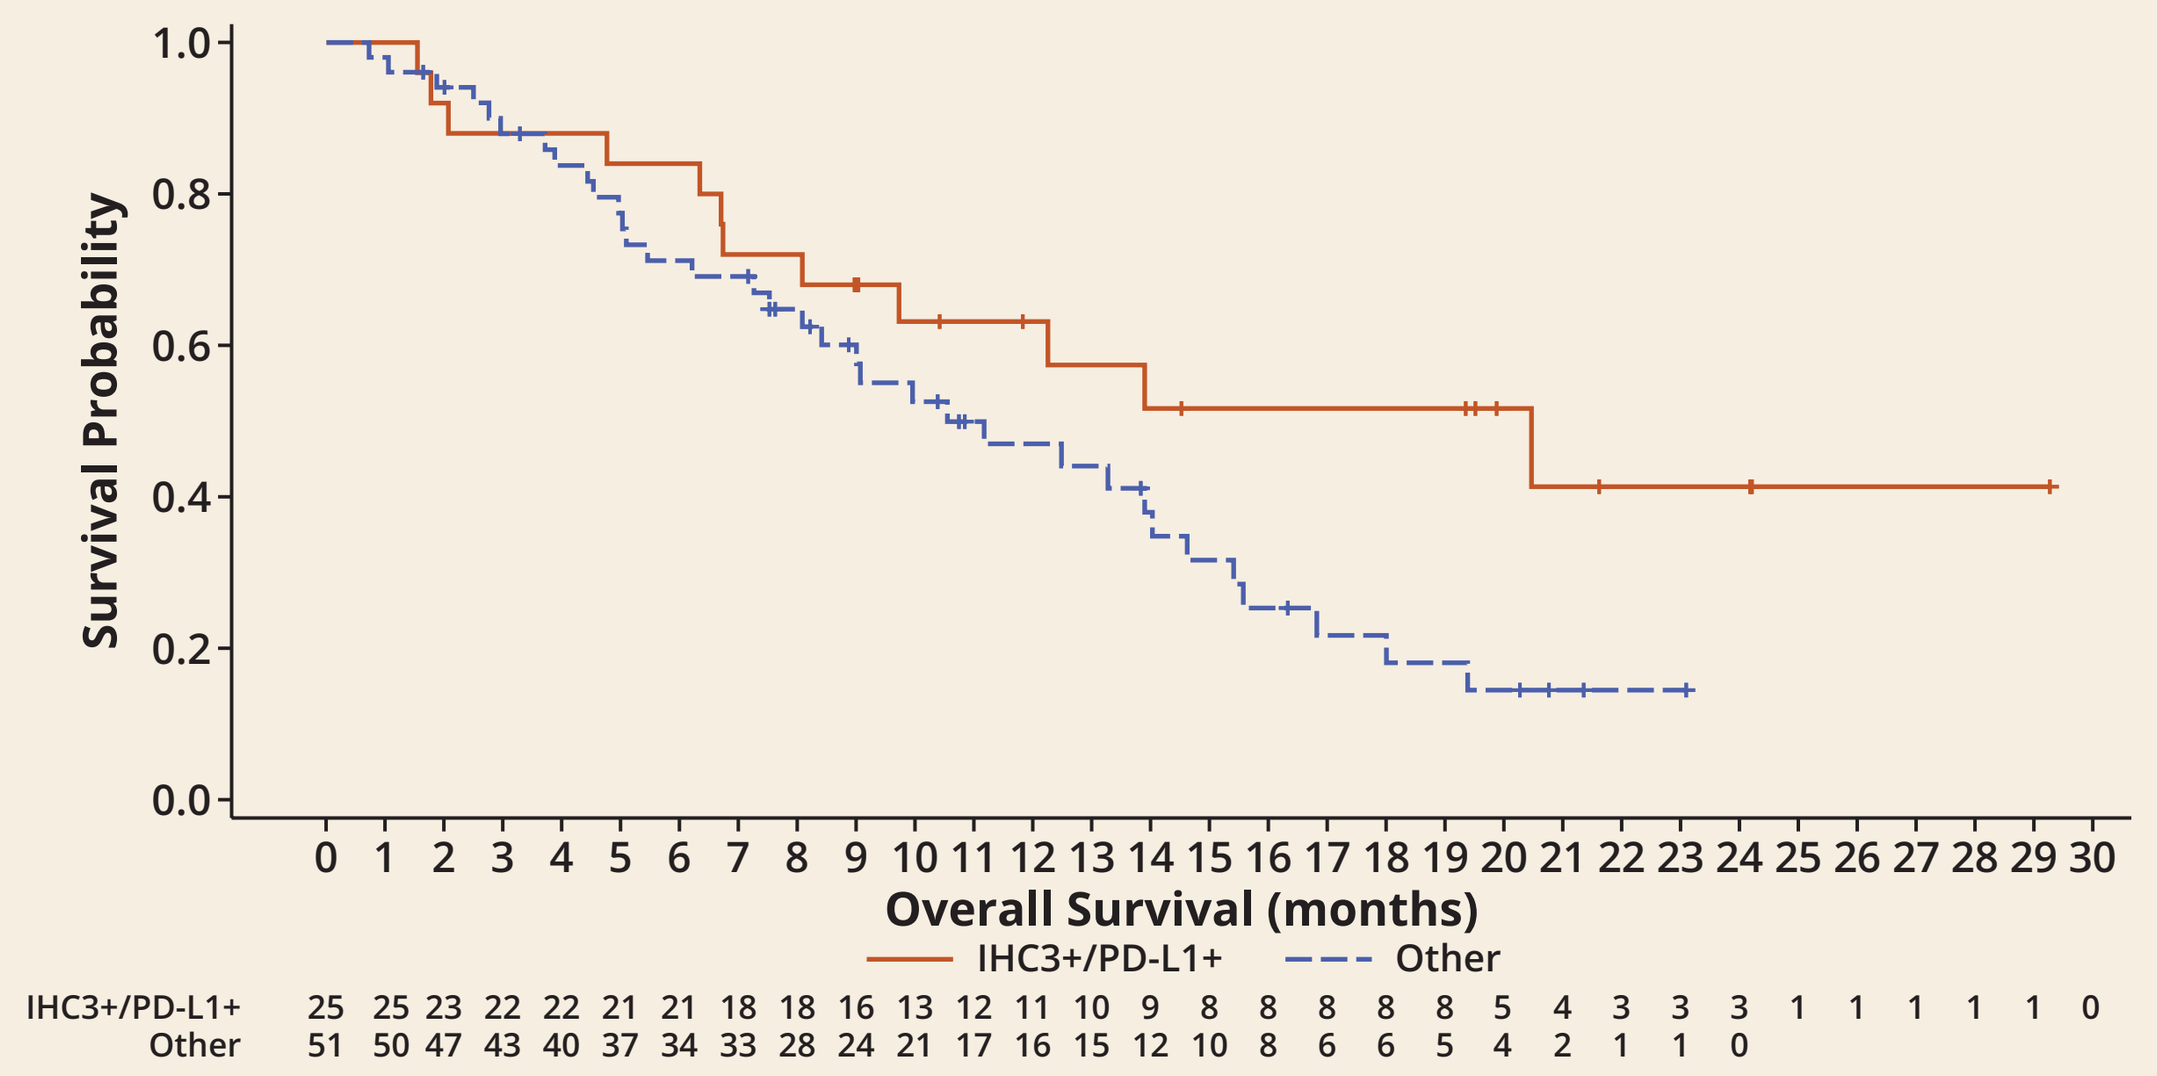

Survival Probability
Overall Survival (months)
IHC3+/PD-L1+
Other
IHC3+/PD-L1+ 25 25 23 22 22 21 21 18 18 16 13 12 11 10 9 8 8 8 8 8 5 4 3 3 3 1 1 1 1 1 0
Other 51 50 47 43 40 37 34 33 28 24 21 17 16 15 12 10 8 6 6 5 4 2 1 1 0

Survival Analysis by HER2 and PD-L1 Expression

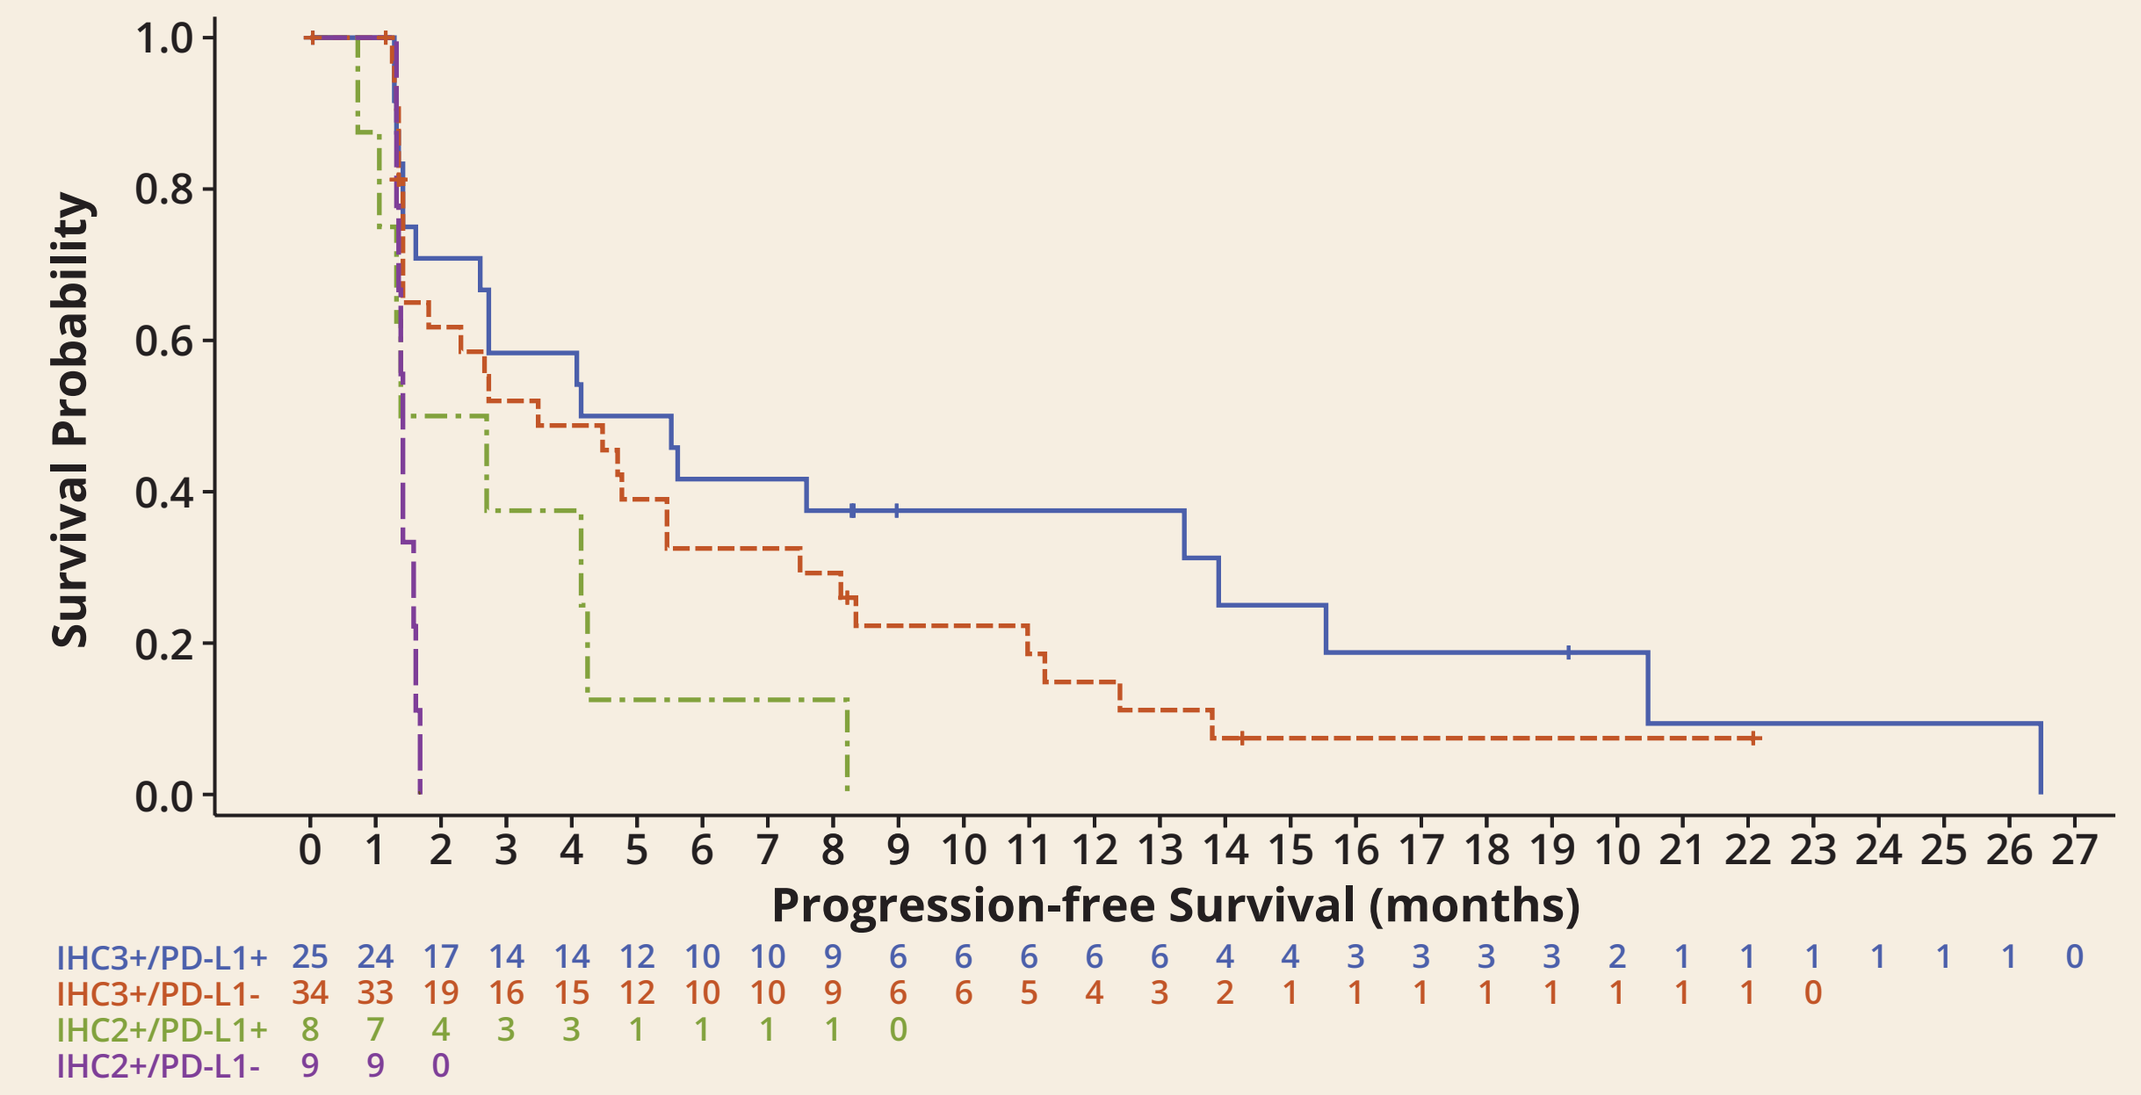

Survival Probability
Progression-free Survival (months)
IHC3+/PD-L1+ 25 24 17 14 14 12 10 10 9 6 6 6 6 6 4 4 3 3 3 3 2 1 1 1 1 1 1 0
IHC3+/PD-L1- 34 33 19 16 15 12 10 10 9 6 6 5 4 3 2 1 1 1 1 1 1 1 1 0
IHC2+/PD-L1+ 8 7 4 3 3 1 1 1 1 0
IHC2+/PD-L1- 9 9 0

Survival Analysis by HER2 and PD-L1 Expression

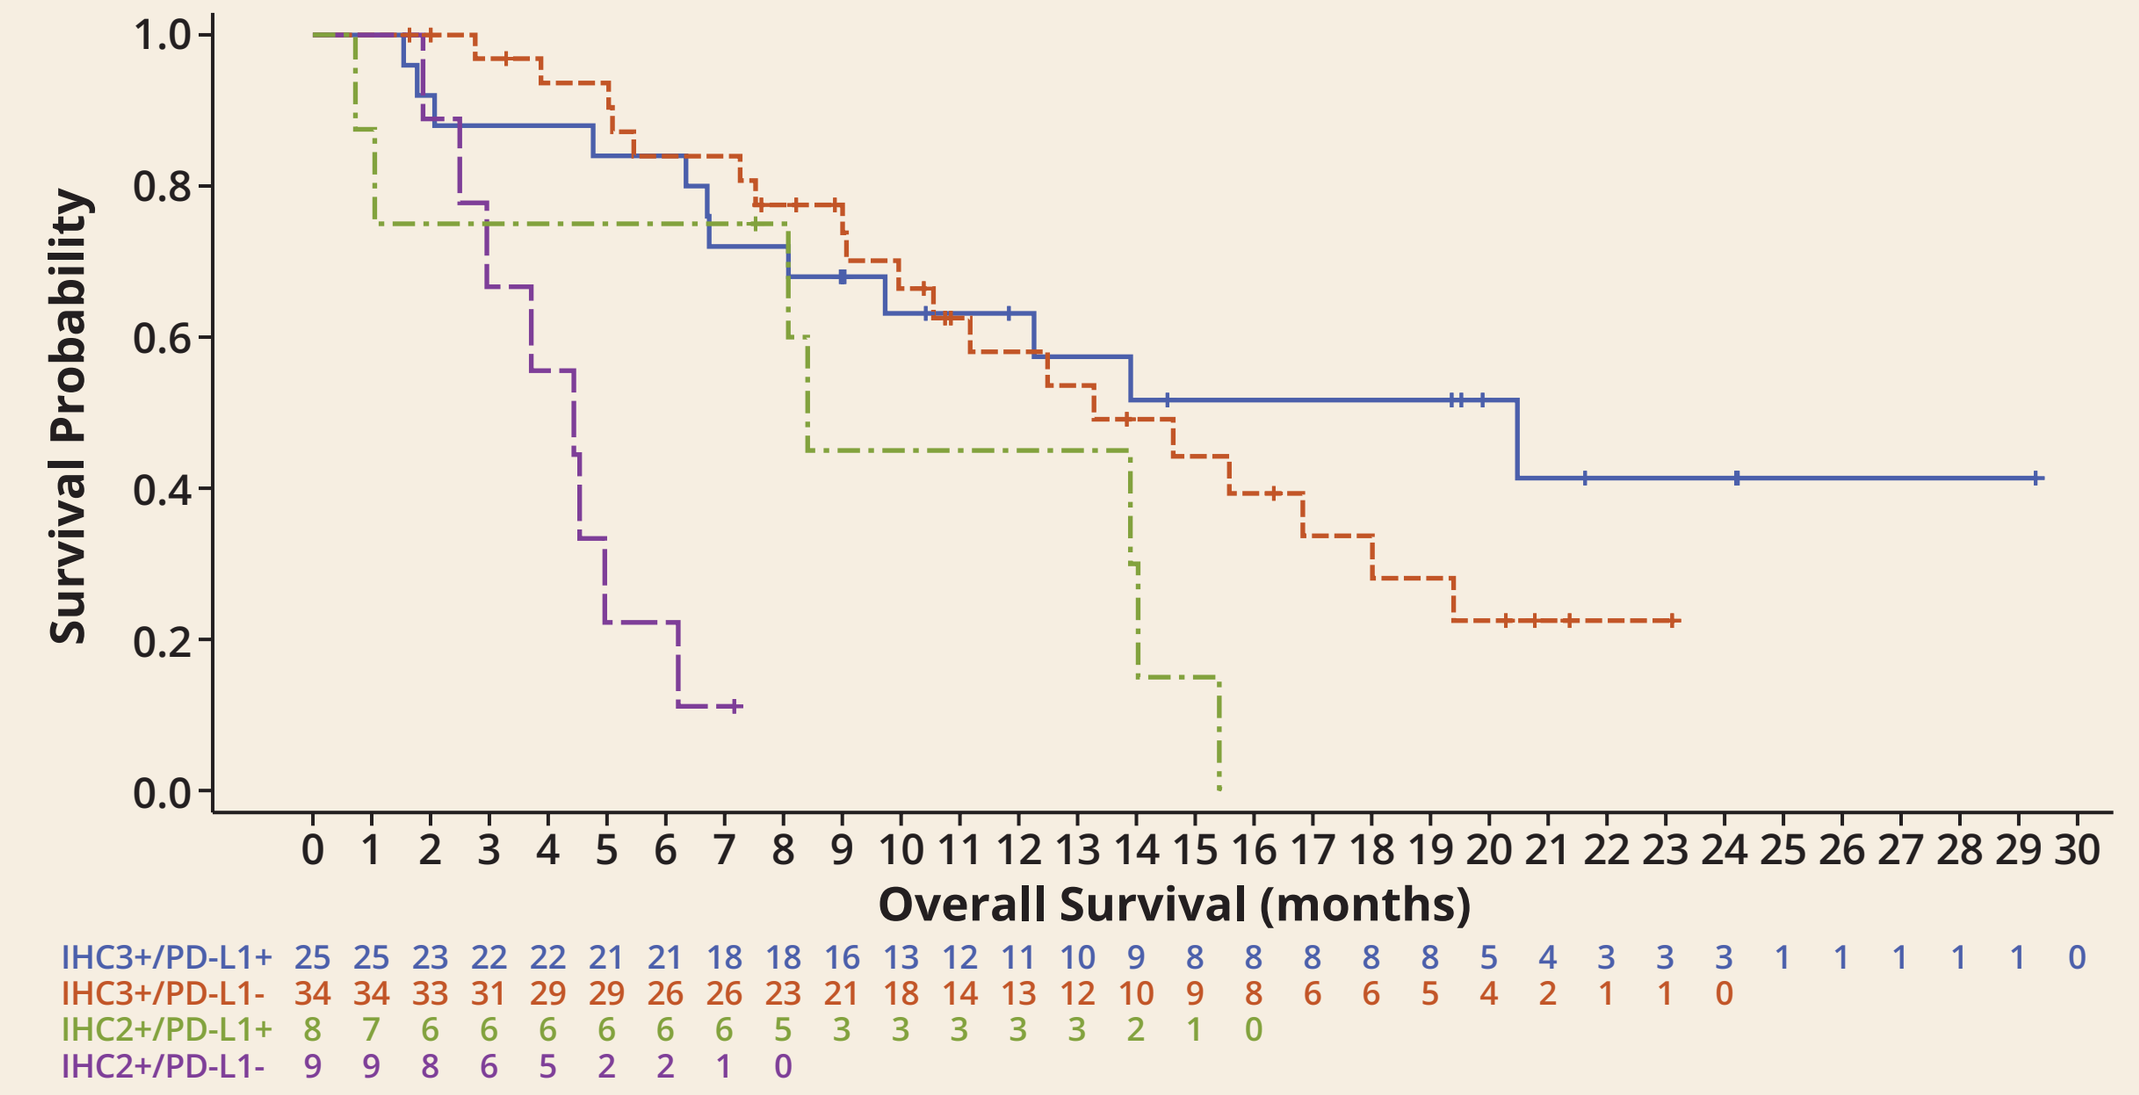

Survival Probability
Overall Survival (months)
IHC3+/PD-L1+ 25 25 23 22 22 21 21 18 18 16 13 12 11 10 9 8 8 8 8 8 5 4 3 3 3 1 1 1 1 1 0
IHC3+/PD-L1- 34 34 33 31 29 29 26 26 23 21 18 14 13 12 10 9 8 6 6 5 4 2 1 1 0
IHC2+/PD-L1+ 8 7 6 6 6 6 6 6 5 3 3 3 3 3 3 2 1 0
IHC2+/PD-L1- 9 9 8 6 5 2 2 1 0

Efficacy Endpoints in Selected Biomarker Positive Populations by Anatomical Site

	Overall ORR (%, n)* DCR (%, n)	Overall mPFS (months) (95% CI)	Overall mOS (months) (95% CI)	Gastric Cancer ORR (%, n) DCR (%, n)	Gastric Cancer mPFS (months) (95% CI)	Gastric Cancer mOS (months) (95% CI)	GEJ Cancer ORR (%, n) DCR (%, n)	GEJ Cancer mPFS (months) (95% CI)	GEJ Cancer mOS (months) (95% CI)
All Patients	(21.74%) 20/92 (54.35%) 50/92	2.73 (1.61, 4.34)	12.48 (9.07, 14.09)	(29.51%) 18/61 (65.57%) 40/61	4.14 (2.60, 5.52)	13.90 (9.72, 20.47)	(6.45%) 2/31 (32.26%) 10/31	1.41 (1.35, 3.61)	9.23 (4.96, 14.03)
IHC3+	(28.17%) 20/71 (63.38%) 45/71	4.34 (2.60, 5.62)	13.90 (10.55, 20.47)	(32.73%) 18/55 (69.09%) 38/55	4.70 (2.66, 7.49)	14.62 (10.55, NR)	(12.50%) 2/16 (43.75%) 7/16	2.63 (1.35, 11.24)	12.01 (5.26, 18.00)
*ERBB2*amp	(33.33%) 16/48 (66.67%) 32/48	4.76 (2.60, 8.11)	13.27 (8.08, 19.38)	(40.00%) 14/35 (77.14%) 27/35	5.49 (2.69, 8.34)	16.82 (9.07, NR)	(15.38%) 2/13 (38.46%) 5/13	1.41 (1.31, 12.39)	5.62 (2.96, 13.27)
PD-L1+	(36.36%) 12/33 (69.70%) 23/33	4.11 (1.41, 7.59)	13.90 (8.08, NR)	(46.15%) 12/26 (80.77%) 21/26	4.19 (2.60, 13.37)	13.90 (8.08, NR)	(0.00%) 0/7 (28.57%) 2/7	1.35 (0.72, 8.21)	14.03 (0.72, NR)
IHC3+/PD-L1+	(48.00%) 12/25 (76.00%) 19/25	4.83 (1.61, 13.90)	20.47 (8.08, NR)	(52.17%) 12/23 (82.61%) 19/23	5.52 (2.60, 13.90)	20.47 (8.08, NR)	(0.00%) 0/2 (0.00%) 0/2	1.28 (NA)	NR (2.07, NR)
*ERBB2*amp/ PD-L1+	(55.56%) 10/18 (83.33%) 15/18	5.52 (1.61, 13.37)	NR (6.74, NR)	(58.82%) 10/17 (88.24%) 15/17	5.52 (1.61, 13.37)	NR (6.74, NR)	(0.00%) 0/1 (0.00%) 0/1	NA	2.07 (NA)
*ERBB2*amp/ IHC3+/PD-L1+	(66.67%) 10/15 (86.67%) 13/15	6.60 (1.61, 15.54)	NR (6.34, NR)	(71.43%) 10/14 (71.43%) 10/14	6.60 (1.61, 15.54)	NR (6.74, NR)	(0.00%) 0/1	NA	2.07 (NA)

*17 confirmed, 3 unconfirmed responses.

Margetuximab + Anti-PD-1 Data in 2nd Line Presents Opportunity to Advance to 1st Line

HER2+ gastric cancer benchmarks

	1st Line SOC	2nd Line SOC	2nd Line Ongoing Phase 2 Study		2nd Line Failed
Agent (Study)	Trastuzumab + Chemo[a] (TOGA)	Ramucirumab + Paclitaxel[b] (RAINBOW)	Margetuximab + Pembrolizumab[c] IHC 3+	Margetuximab + Pembrolizumab[c] IHC 3+/PD-L1+	Pembrolizumab[d] (KEYNOTE-61) ✖
ORR	47%	28%	33%	52%	15.8% (PD-L1+)
Median PFS	6.7 mos.	4.4 mos.	4.7 mos.	5.5 mos.	1.5 mos.
Median OS	**13.1 mos.**	9.6 mos.	**14.6 mos.**	**20.5 mos.**	9.1 mos
≥ Grade 3 TRAEs	**68%**	Overall: N/A 41% Neutropenia 15% Hypertension 12% Fatigue	**20%**	**20%**	14.3%
Gastric/GEJ Patient Mix	80/20%	80/20%	100%/0%	100%/0%	Not disclosed

[a]Data from Herceptin package insert; Bang, et al., *Lancet*, 2010. [b]Data from Cyramza package insert; Wilkes, et al., *Lancet Oncology*, 2014. [c]Grade 3 TRAE includes all GC and GEJ patients (n=92). [d]Data presented at ASCO 2018, Abstract 4062.

Conclusions

- Margetuximab is an Fc-engineered anti-HER2 antibody that mediates enhanced innate responses and leads to increased HER2-specific adaptive immune responses in patients with HER2+ gastric and breast carcinoma
- In this study, the combination of margetuximab + pembrolizumab, as a chemotherapy-free regimen, in patients with HER-2+ GEA that have progressed/recurred after front-line therapy including trastuzumab has shown:
 - An acceptable safety and tolerability
 - A prolonged median OS (12.9 mos) compared to historical experience with 2L standard of care (RAINBOW, ramucirimab + taxane, mOS=9.6 mos) or checkpoint inhibitor alone (KN-061, pembrolizumab, mOS=9.1 mos)
- In the HER2 IHC3+/PD-L1+ gastric cancer patients a prolonged median OS (20.5 mos) was noted, which exceeds historical experience with 1L standard of care (TOGA, trastuzumab + chemo, mOS=13.1 mos)
- These results are particularly notable because many patients lose HER2 expression post trastuzumab.
- Based on these observations, the combination of margetuximab + a checkpoint inhibitor could provide a potential chemotherapy-free regimen for the treatment of GEA and/or be used with chemotherapy to improve the clinical activity of existing 1L SoC
- A Phase 2/3 study (MAHOGANY) is being initiated to evaluate margetuximab in combination with a checkpoint inhibitor with or without chemotherapy in 1L GEA

MAHOGANY Phase 2/3 Study: Registration Path in 1L Gastric and GEJ Cancer

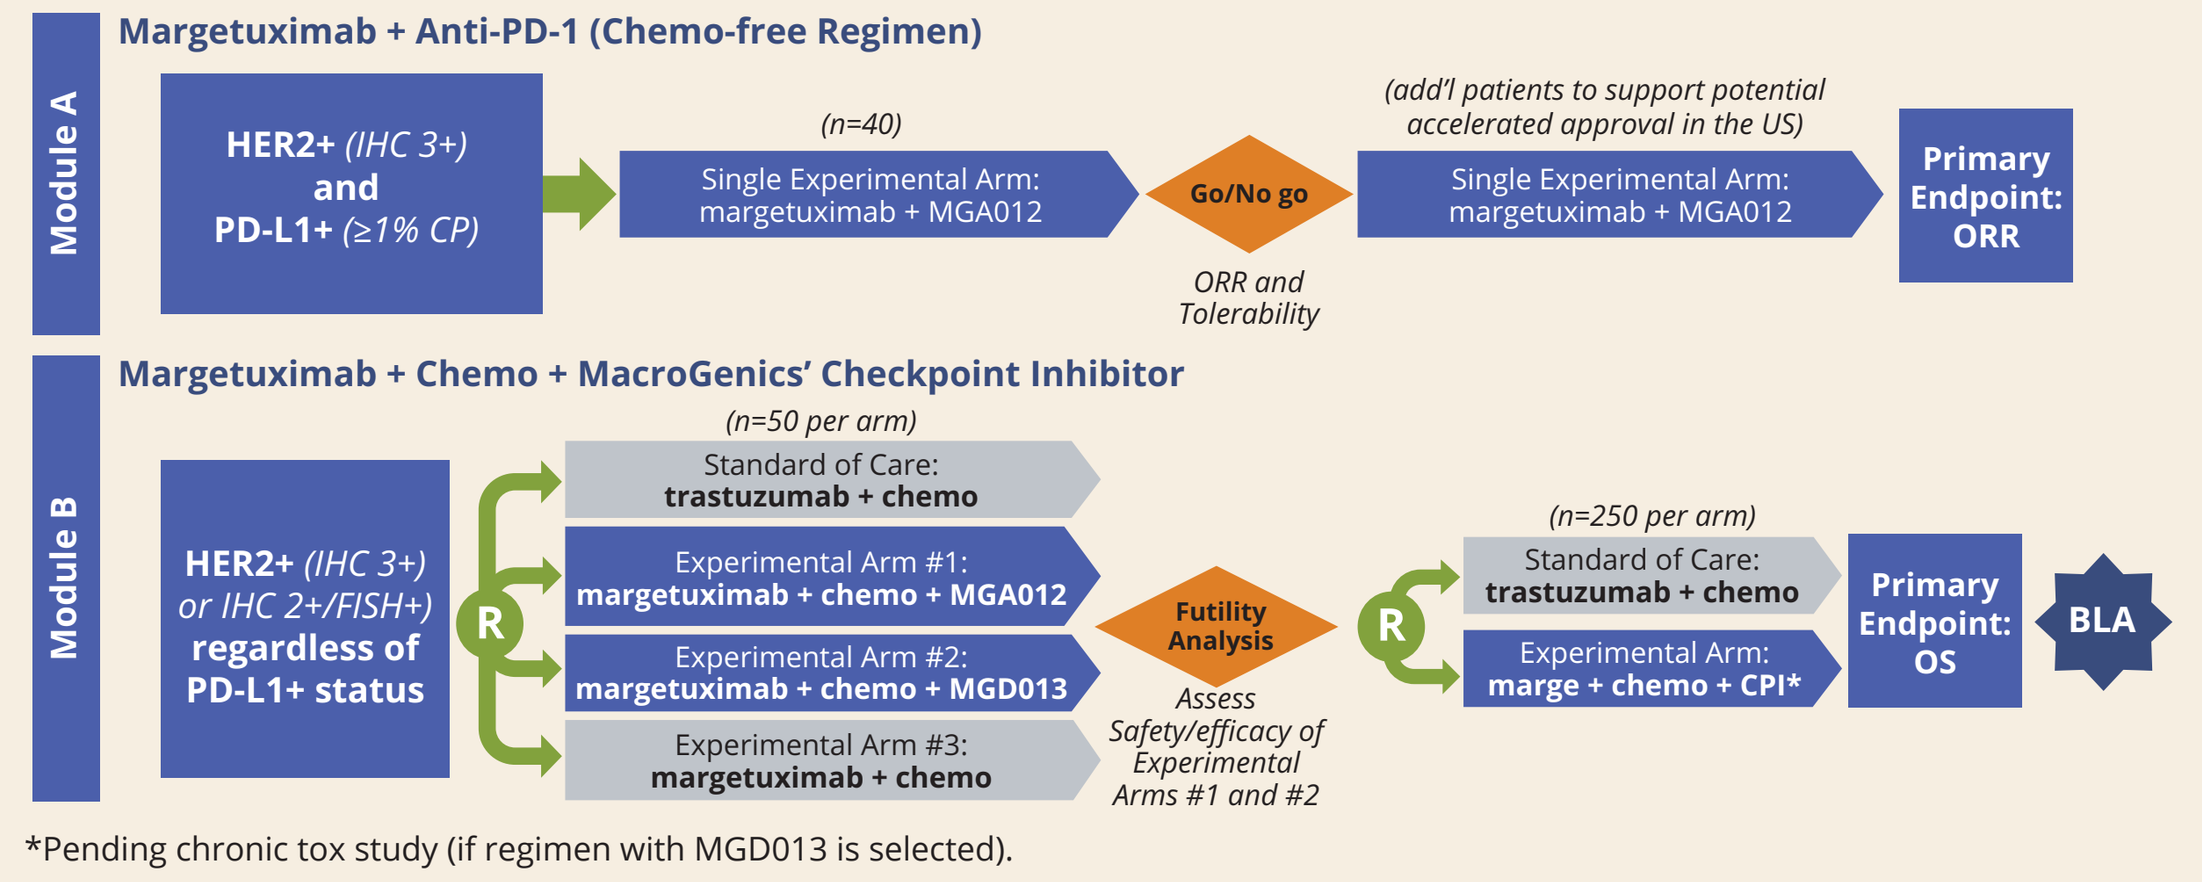

Module A
Margetuximab + Anti-PD-1 (Chemo-free Regimen)
HER2+ (IHC 3+) and PD-L1+ (≥1% CP)
(n=40)
Single Experimental Arm: margetuximab + MGA012
Go/No go
ORR and Tolerability
(add'l patients to support potential accelerated approval in the US)
Single Experimental Arm: margetuximab + MGA012
Primary Endpoint: ORR
Module B
Margetuximab + Chemo + MacroGenics' Checkpoint Inhibitor
HER2+ (IHC 3+) or IHC 2+/FISH+) regardless of PD-L1+ status
R
(n=50 per arm)
Standard of Care: trastuzumab + chemo
Experimental Arm #1: margetuximab + chemo + MGA012
Experimental Arm #2: margetuximab + chemo + MGD013
Experimental Arm #3: margetuximab + chemo
Futility Analysis
Assess Safety/efficacy of Experimental Arms #1 and #2
(n=250 per arm)
Standard of Care: trastuzumab + chemo
Experimental Arm: marge + chemo + CPI*
Primary Endpoint: OS
BLA

*Pending chronic tox study (if regimen with MGD013 is selected).

This study was sponsored by MacroGenics, Inc. Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented at the 2019 Annual Congress of the European Society for Medical Oncology, September 27–October 1, 2019, Barcelona, Spain

davidsonj@macrogenics.com